52.24

Neuroprotection of Hippocampal CA1 Neurons from Ischemic Cell Death Using the Calcium Binding Protein Aequorin

Julia A. Detert[1], John D. Heisler[1], Erin L. Hochstetter[1], Trevor M. Van Langendon[1], & James R. Moyer, Jr.[1,2]

Departments of [1]Psychology and [2]Biological Sciences

ABSTRACT

During ischemia, the deprivation of blood flow and oxygen to the brain results in excessive calcium influx through glutamate receptors, which can rapidly trigger cell death. One way neurons protect themselves from the toxic effects of calcium is to buffer the calcium with calcium binding proteins (CaBPs). Previous work has demonstrated that hippocampal neurons expressing the CaBP calbindin-D28k are better able to withstand an excitotoxic insult than neurons lacking calbindin. We have been investigating the feasibility of regulating calcium levels during ischemia by replenishing CaBPs. Aequorin (AQ) is a 22 kDa CaBP isolated from the coelenterate *Aequorea victoria.* AQ has been used for years as an auto-fluorescent indicator for monitoring calcium levels and has been shown to be safe and well tolerated by cells. The present studies were designed to test the hypothesis that intrahippocampal infusion of AQ can protect neurons from an ischemic insult. Rats were stereotaxically implanted with bilateral cannula (in the CA1 region of the dorsal hippocampus) under aseptic conditions. After recovery, rats received an intrahippocampal infusion of AQ (0.4%, 1%, or 4%) in one hemisphere and artificial CSF (aCSF) in the other (0.5 µl/min for 1 min). Twenty-four or 72 hours following the infusion, coronal brain slices (400 µm) were cut with a vibratome. Slices were maintained in oxygenated aCSF for 1 hr. They were then subjected to a 5-min oxygen-glucose deprivation (OGD), returned to oxygenated aCSF (with 0.2% trypan blue) for a 30-min reperfusion and then rinsed in oxygenated aCSF. All slice experiments were carried out at 35 °C. Slices were then fixed, cryoprotected, sub-sectioned (40 µm), mounted, and coverslipped. An individual blind to treatment group counted the number of trypan blue stained (dead) CA1 neurons, and the number of dead cells in the AQ-treated hemisphere was compared to the aCSF-treated hemisphere to calculate a percent rescue. AQ treatment prior to OGD resulted in significantly fewer trypan blue stained CA1 neurons relative to control. In addition, the rats injected with 4% AQ had more rescue (58 ± 12%) than those injected with 0.4% AQ (37 ± 20%). However, when OGD was initiated 72 hours after 4% AQ infusion, no neuroprotection was noted. We are currently evaluating other time points to determine the time course over which AQ is neuroprotective. These data support the hypothesis that AQ may be an effective neurotherapeutic against ischemia when administered within 24 hours prior to an ischemic insult. We are also in the process of determining whether delivery of AQ is neuroprotective when administered *following* an ischemic insult.

INTRODUCTION

- Neurons are continuously subjected to elevations in intracellular Ca^{2+} as a result of ongoing activity and this elevation is necessary for certain normal neuronal processes to occur, however too much Ca^{2+} can be toxic (Bano et al., 2005; Choi, 1992; Lee et al., 1999).
- As a result, the intracellular Ca^{2+} concentration in neurons is very tightly regulated (Kristian & Siesjo, 1998).
- Several mechanisms enable neurons to limit or control cytosolic Ca^{2+} (Baimbridge et al., 1992; Chard et al., 1993), including calcium binding proteins (CaBPs).
- The presence of CaBPs confers some protection against excitotoxic insults, which would normally kill the cell (Gary et al., 2000).
- Decreased levels of CaBPs are observed with advancing age (De Jong et al., 1996; Krzywkowski et al., 1996; Moyer et al., in press; Villa et al.,1994), and in neurodegenerative disorders (Mattson & Magnus, 2006), including:
 - Alzheimer's disease (Hof & Morrison, 1991; Iacopino & Christakos, 1990; Mikkonen et al., 1999; Sutherland et al., 1993)
 - Parkinson's disease (Iacopino & Christakos, 1990)
 - Ischemia (Yenari et al., 2001).
- During ischemia, neurons are subjected to excess Ca^{2+} influx triggering a cascade of events leading to cell death (Choi, 1992).
- Since neuronal CaBPs are depleted in neurodegenerative disorders, and since neurons that express CaBPs are better able to survive an excitotoxic challenge, we reasoned that supplementing with CaBPs prior to an ischemic insult might be neuroprotective.
- Treatments aimed at minimizing Ca^{2+} toxicity during ischemia have been administered before an ischemic insult, with positive results.
 - Yenari et al. (2001) treated animals with calbindin prior to ischemia and found that overexpression of calbindin resulted in fewer dead neurons.
 - Fan et al. (2007) found a smaller infarct volume, better behavioral recovery, and decreased apoptosis in rats pre-treated with calbindin.
- Aequorin (AQ) is a 22 kDa CaBP isolated from the coelenterate *Aequorea victoria.*

Hypothesis: supplementing CaBPs with AQ in the hippocampus will be neuroprotective when administered prior to an ischemic insult.

METHODS

Animals. Fifty-three male F344 adult rats (mean age = 4.6 ± 0.1 mo.) were used. Rats were kept on a 14/10-hour day/night cycle with free access to food and water.
Surgery. Rats were anesthetized and mounted on a stereotaxic apparatus. Under aseptic conditions, the scalp was incised and retracted to the side, and the head was leveled between bregma and lambda. Each rat was prepared with bilateral stainless steel guide cannulae aimed at the dorsal hippocampus (dhpc) using sterotaxic coordinates (3.5 mm posterior, ± 2.6 mm lateral, 3.0 mm ventral) relative to bregma. Cannulae were secured to the skull with stainless steel screws and epoxy. A stainless steel cap remained in place when the rats were not being injected to prevent the guide cannulae from becoming occluded.
Drugs and Infusions. Rats were given an infusion of 0, 0.4, 1, or 4% AQ in one hemisphere and aCSF in the other hemisphere 1, 24, 48, or 72 hours prior to decapitation. To facilitate neuronal uptake of AQ, 6% DMSO was added. All rats received bilateral infusions (0.5 µl/side) over 60 seconds and the injection cannulae remained in place for an additional 2 min to ensure diffusion. The infusion cannulae were cut to extend 0.5 mm beyond the guide cannulae.
Slice Preparation. 400 µm thick slices were prepared using standard procedures (Moyer & Brown, 1998). Following slice recovery, *in vitro* ischemia was induced by transferring slices to fructose-CSF (glucose replaced with fructose and bubbled with 95% N_2 – 5% CO_2 instead of a 95% O_2 – 5% CO_2). The slices were in the ischemia condition for 5 minutes, and were then returned to oxygenated aCSF that contained 0.2% trypan blue for 30 minutes. Trypan blue readily penetrates dead cells and stains them blue while leaving living cells unstained (DeRenzis & Schechtman, 1973). The slices were rinsed in oxygenated room temperature aCSF twice then fixed in 10% neutral buffered formalin overnight in the refrigerator. Slices were then cryoprotected, cut on a cryostat (40 µm), and mounted onto subbed slides.
Post-ischemia AQ. Slices were prepared as listed above and were transferred to interface chambers for one hour of recovery. *In vitro* ischemia was induced by switching the perfusion solution to fructose-CSF bubbled with 95% N_2 – 5% CO_2 and the chambers were filled with 95% N_2 – 5% CO_2 gas for 5 minutes. Immediately following *in vitro* ischemia, perfusion solution and chambers were returned to oxygenated aCSF and 4% AQ or aCSF (25 µL) was pipetted onto each slice. Five minutes later, the slices were removed from the interface chambers and stained with 0.2% trypan blue as above.
Cell Counts. Slices were examined under an Olympus microscope (equipped with a digital camera) at 10X, and pictures were taken. Trypan blue stained neurons within CA1 (about an 800 µm section) were counted by an experimenter blind to experimental conditions. Statistical analyses were performed using Statview (v 5.0; SAS Institute, Inc., Cary, NC). An ANOVA was used to evaluate a drug effect. Asterisk indicates $p < .05$ from 0% AQ.
Western Blots. Rats were given a bilateral injection of AQ and sacrificed at one of the following time points: 1 hr, 24 hr, 48 hr, or 72 hr. Brains were removed, rapidly frozen, and stored at -80°C. The dhpc and ventral hippocampus (vhpc) were dissected out and homogenized separately. Samples were centrifuged and the supernatant removed and measured using a Bradford protein assay kit (Bio-Rad). Protein samples were normalized and loaded for SDS-PAGE (9%). Proteins were transferred onto membranes using a semidry transfer apparatus (Bio-Rad). Membranes were then incubated in blocking buffer (2 hr), primary antibody (overnight at 4 °C; 1:200 mouse anti-aequorin [Chemicon], and secondary antibody (90 min; 1:5000 anti-mouse [Santa Cruz Biotechnology]). Membranes were then washed, placed in a chemiluminescence solution (Santa Cruz Biotechnology), and exposed to autoradiagraphic film (Hyperfilm MP). Images were taken and densitometry was performed using NIH Image J Software by an experimenter blind to lane conditions. A percentage of control score was derived for each rat by dividing each animal's relative optical density score by the mean of the 1 hr time point.

AEQUORIN IS NEUROPROTECTIVE

1. Aequorin injected prior to ischemia is neuroprotective

A. Control

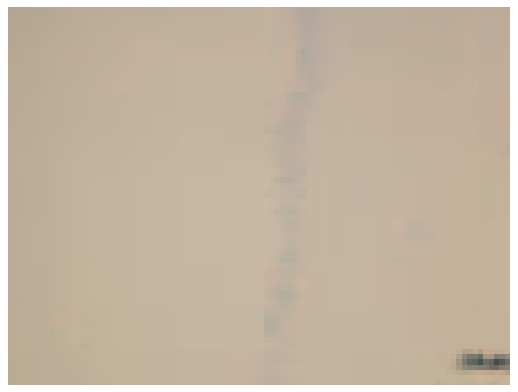

B. 4% Aequorin

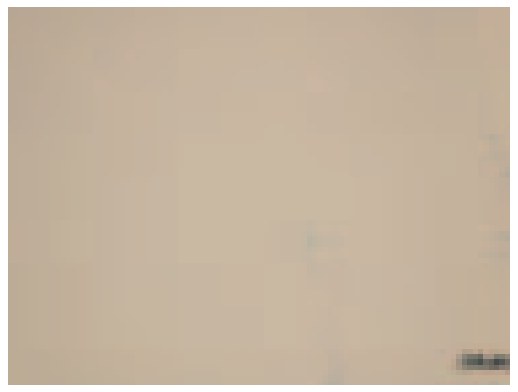

C. Effectiveness of different doses of AQ

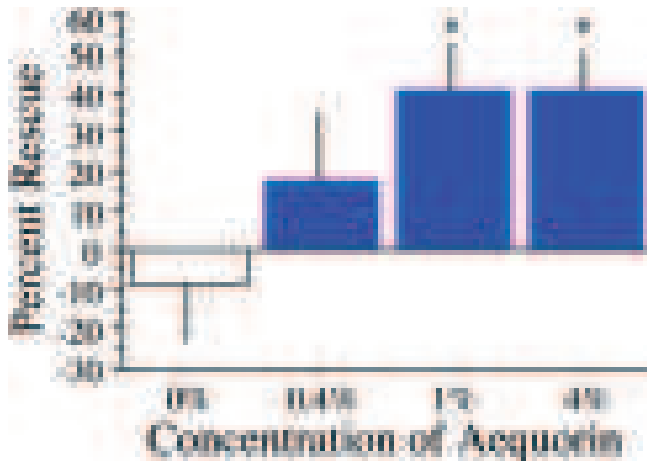
Percent Rescue
60
50
40
30
20
10
0
-10
-20
-30
0%
0.4%
1%
4%
Concentration of Aequorin

TIME-DEPENDENT EFFECT OF AEQUORIN

2. Neuroprotective effects of aequorin last less than 48 hours

A. Window of neuroprotection

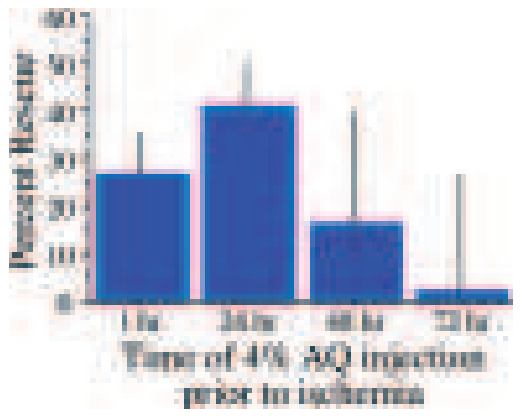
Percent Rescue
1 hr
24 hr
48 hr
72 hr
Time of 4% AQ injection prior to ischemia

B. AQ 48 hr prior to ischemia

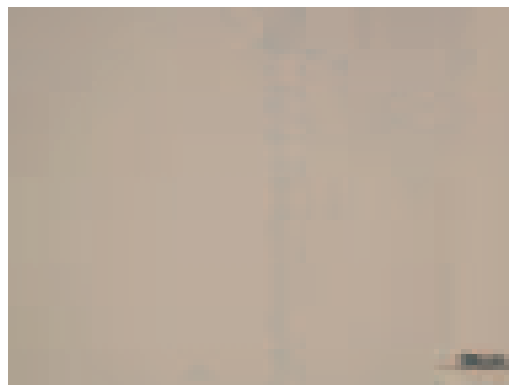

3. Aequorin is detected in hippocampus at 24 hours, but not 48 hrs

A. Time course of AQ in dhpc

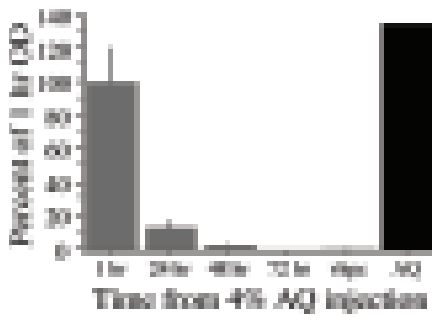
Percent of 1 hr OD
1 hr
24 hr
48 hr
72 hr
vhpc
AQ
Time from 4% AQ injection

B. Representative blots

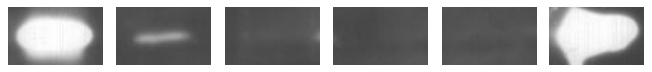
22 kDa
1 hr
24 hr
48 hr
72 hr
vhpc
AQ

POST-ISCHEMIA AEQUORIN

4. Aequorin administered immediately after ischemia is neuroprotective

A. Control

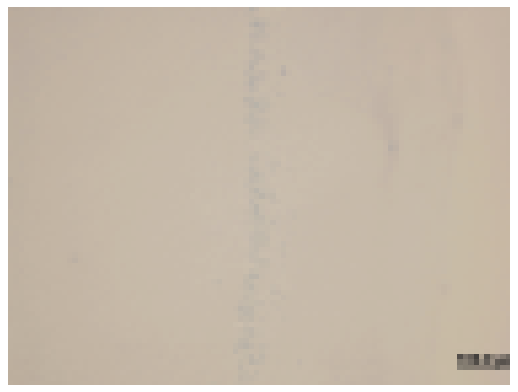

B. Aequorin

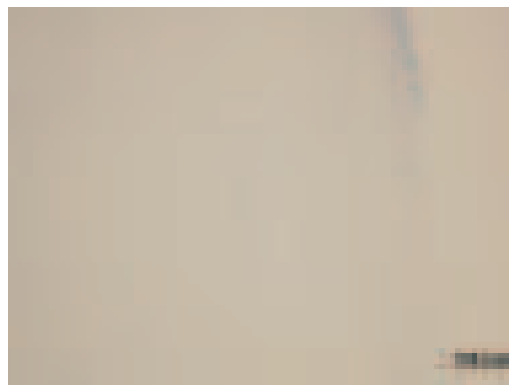

SUMMARY & IMPLICATIONS

1. **The calcium binding protein aequorin is neuroprotective when injected into hippocampus prior to ischemia**
 - *There were fewer dead cells in area CA1 of the hippocampus in the aequorin-injected hemisphere compared to the control-injected hemisphere*
2. **The neuroprotective effect of aequorin is time-dependent**
 - *When injected 48 or 72 hrs prior to ischemia, aequorin was no longer neuroprotective*
3. **Aequorin is neuroprotective when administered post-ischemia**
 - *Preliminary data show there were fewer dead cells in slices given aequorin immediately after 5 min OGD compared to slices given aCSF*

REFERENCES

Baimbridge, K. G., Celio, M. R., & Rogers, J. H. (1992). Calcium-binding proteins in the nervous system. *Trends in Neuroscience, 15*(8), 303-308
Bano, D., Young, K. W., Guerin, C. J., Lefeuvre, R., Rothwell, N. J., Naldini, L., et al. (2005). Cleavage of the plasma membrane Na^+/Ca^{2+} exchanger in excitotoxicity. *Cell, 120*(2), 275-285.
Chard, P. S., Bleakman, D., Christakos, S., Fullmer, C. S., & Miller, R. J. (1993). Calcium buffering properties of calbindin D28k and parvalbumin in rat sensory neurones. *Journal of Physiology, 472*, 341-357.
Choi, D. W. (1992). Excitotoxic cell death. *Journal of Neurobiology, 23*(9), 1261-1276.
DeRenzis, F. A., & Schechtman, A. (1973). Staining by neutral red and trypan blue in sequence for assaying vital and nonvital cultured cells. *Stain Technology, 48*(3), 135-136.
De Jong, G. I., Naber, P. A., Van der Zee, E. A., Thompson, L. T., Disterhoft, J. F., & Luiten, P. G. M. (1996). Age-related loss of calcium binding proteins in rabbit hippocampus. *Neurobiology of Aging, 17*(3), 459-465.
Fan, Y., Shi, L., Gu, Y., Zhao, Y., Xie, J., Qiao, J., et al. (2007). Pretreatment with PTD-calbindin D 28k alleviates rat brain injury induced by ischemia and reperfusion. *Journal of Cerebral Blood Flow and Metabolism, 27*(4), 719-728.
Gary, D. S., Sooy, K., Chan, S. L., Christakos, S., & Mattson, M. P. (2000). Concentration- and cell type-specific effects of calbindin D28k on vulnerability of hippocampal neurons to seizure-induced injury. *Brain Research. Molecular Brain Research, 75*(1), 89-95.
Hof, P. R., & Morrison, J. H. (1991). Neocortical neuronal subpopulations labeled by a monoclonal antibody to calbindin exhibit differential vulnerability in Alzheimer's disease. *Experimental Neurology, 111*, 293-301.
Iacopino, A. M., & Christakos, S. (1990). Specific reduction of calcium-binding protein (28-kilodalton calbindin-D) gene expression in aging and neurodegenerative diseases. *Proceedings of the National Academy of Sciences (USA), 87*, 4078-4082.
Kristian, T., & Siesjo, B. K. (1998). Calcium in ischemic cell death. *Stroke, 29*(3), 705-718.
Krzywkowski, P., Potier, B., Billard, J. M., Dutar, P., & Lamour, Y. (1996). Synaptic mechanisms and calcium binding proteins in the aged rat brain. *Life Sciences, 59*(5/6), 421-428.
Lee, J. M., Zipfel, G. J., & Choi, D. W. (1999). The changing landscape of ischaemic brain injury mechanisms. *Nature, 399*(6738 Suppl), A7-14.
Mattson, M. P., & Magnus, T. (2006). Ageing and neuronal vulnerability. *Nature Reviews. Neuroscience, 7*(4), 278-294.
Mikkonen, M., Alafuzoff, I., Tapiola, T., Soininen, H., & Niettinen, R. (1999). Subfield- and layer-specific changes in parvalbumin, calretinin and calbindin-D28K immunoreactivity in the entorhinal cortex in Alzheimer's disease. *Neuroscience, 92*(2), 515-532.
Moyer, J. R., Jr., & Brown, T. H. (1998). Methods for whole-cell recording from visually preselected neurons of perirhinal cortex in brain slices from young and aging rats. *Journal of Neuroscience Methods, 86*(1), 35-54.
Moyer, J. R., Jr., Furtak, S. C., McGann, J. P., & Brown, T. H. (in press). Aging-related changes in calcium binding proteins in rat perirhinal cortex. *Neurobiology of Aging*.
Sutherland, M. K., Wong, L., Somerville, M. J., Yoong, L. K. K., Bergeron, C., Parmentier, M., et al. (1993). Reduction of calbindin-28k mRNA levels in Alzheimer as compared to Huntington hippocampus. *Brain Research. Molecular Brain Research, 18*(1-2), 32-42.
Villa, A., Podini, P., Panzeri, M. C., Racchetti, G., & Meldolesi, J. (1994). Cytosolic Ca^{2+} binding proteins during rat brain ageing: loss of calbindin and calretinin in the hippocampus, with no change in the cerebellum. *European Journal of Neuroscience, 6*, 1491-1499.
Yenari, M. A., Minami, M., Sun, G. H., Meier, T. J., Kunis, D. M., McLaughlin, J. R., et al. (2001). Calbindin d28k overexpression protects striatal neurons from transient focal cerebral ischemia. *Stroke, 32*(4), 1028-1035.

Research Supported by Quincy Bioscience, LLC