Neuroprotective Effects of Aequorin on Hippocampal CA1 Neurons Following Ischemia

Julia A. Detert[1], Patrick K. Tao[1], Liviu Bunaciu[1], Melody L. Schmidt[2], &
James R. Moyer, Jr.[1,2]
Departments of [1]Psychology and [2]Biological Sciences
University of Wisconsin-Milwaukee, Milwaukee, WI 53201

ABSTRACT

Calcium plays a pivotal role in various neuronal signaling cascades, including neurotransmitter release, enzyme activation, gene expression, and synaptic plasticity. However, during ischemia, excessive calcium influx through glutamate receptors can rapidly trigger cell death. Calcium binding proteins (CaBPs) may serve an important role in buffering neurons from potentially toxic elevations in the concentration of intracellular calcium. Previous work has demonstrated that hippocampal neurons expressing the CaBP calbindin-D28k are better able to withstand an excitotoxic insult than neurons lacking calbindin. Since calbindin levels are depleted very early during the aging process (Moyer et al., 2001), we have been investigating the feasibility of replenishing CaBPs as a neurotherapeutic. Aequorin (AQ) is a 22 kDa CaBP isolated from the coelenterate *Aequorea Victoria*. AQ has been used for years as a luminescent indicator for monitoring calcium levels and has been shown to be safe and well tolerated by cells. The present studies were designed to test the hypothesis that intrahippocampal injection of AQ can protect neurons from an ischemic insult. Rats were stereotaxically implanted with bilateral cannula (in the CA1 region of the dorsal hippocampus) under aseptic conditions. After recovery, each rat received inrahippocampal injection of 4% AQ or artificial CSF (aCSF; 0.5 µl/min for 1 min). Twenty-four hours following the injections, coronal brain slices (400 µm) were cut with a vibratome (Moyer et al., 2002). Slices were kept in oxygenated aCSF for 1 hr and then subjected to a 5 min ischemic insult (i.e., glucose was replaced by fructose; oxygen was replaced by nitrogen). Slices were returned to oxygenated aCSF (with 0.2% trypan blue) for 30 min (35 °C for all experiments). Slices were rinsed in oxygenated aCSF, fixed, transferred to 30% sucrose, sectioned on a cryostat (40 µm), mounted onto slides, and coverslipped. The number of trypan blue stained CA1 neurons was counted. Five experiments demonstrated that AQ treatment resulted in significantly fewer trypan blue stained CA1 neurons relative to controls (AQ: 57 ± 11; aCSF: 117 ± 14; $F(1,13) = 11.2$, $p < .01$). These data support the hypothesis that AQ may be an effective neurotherapeutic against ischemia. Future studies will determine how long AQ remains in neurons and whether delivery of AQ is neuroprotective when administered after an ischemic insult.

INTRODUCTION

During normal aging as well as neurodegenerative disorders, impaired Ca^{2+} homeostasis may play a critical role in cellular aging processes (Gant, Sama, Landfield, & Thibault, 2006). Calcium plays a pivotal role in various neuronal processes, including neurotransmitter release (Lin & Scheller, 2000), gene expression, and synaptic plasticity (West et al., 2001). Neurons are continuously subjected to elevations in intracellular calcium as a result of ongoing activity and this elevation is necessary for certain normal neuronal processes to occur, however too much calcium can be toxic (Bano et al., 2005; Choi, 1992, 2005; Lee, Zipfel, & Choi, 1999). Thus, in neurons, the intracellular calcium concentration is very tightly regulated (Kristian & Siesjo, 1998). Several mechanisms enable neurons to limit or control cytosolic calcium (Baimbridge, Celio, & Rogers, 1992; Chard, Bleakman, Christakos, Fullmer, & Miller, 1993). In particular, calcium binding proteins (CaBPs; such as calbindin, parvalbumin, and calretinin) are particularly important for binding and buffering cytosolic calcium .

Studies in the hippocampus (HPC) have shown that the presence of CaBPs confers some protection against excitotoxic insults, which would normally kill the cell (Gary, Sooy, Chan, Christakos, & Mattson, 2000). Furthermore, decreased levels of CaBPs are observed with advancing age (De Jong et al.,1996; Krzywkowski, Potier, Billard, Dutar, & Lamour, 1996; Villa, Podini, Panzeri, Racchetti, & Meldolesi, 1994), and in neurodegenerative disorders (Mattson & Magnus, 2006), including Alzheimer's disease (Hof & Morrison, 1991; Iacopino & Christakos, 1990; Mikkonen, Alafuzoff, Tapiola, Soininen, & Niettinen, 1999; Sutherland et al., 1993), Parkinson's disease (Iacopino & Christakos, 1990), and stroke (Yenari et al., 2001).

During ischemia neurons are subjected to excess calcium influx triggering a cascade of events leading to cell death (Choi, 1992). Since neuronal CaBPs are depleted in neurodegenerative disorders, and since neurons that express CaBPs are better able to survive an excitotoxic challenge, we reasoned that replenishing depleted CaBPs might provide a neuroprotective function. Furthermore, data from rodent studies suggest that aging-related decreases in CaBPs begin during middle age (Villa et al., 1994), when animals are only beginning to show learning and memory deficits (Moyer & Brown, 2006). One possible explanation is that the decrease in neuronal CaBPs that occurs in middle-aged animals may ultimately leave those neurons more vulnerable to neurodegeneration in old age.

Treatments aimed at minimizing the effects of ischemia on cell death have been administered before or after an ischemic insult, with positive results. For example, Yenari et al (2001) treated animals with calbindin prior to inducing ischemia and found that overexpression of calbindin resulted in fewer dead neurons. Another approach that has been shown to help prevent ischemia is to improve blood flow (Grotta, 1987). In addition, neuroprotective effects have been observed when glutamate receptor antagonists were administered following an ischemic event (Makarewicz, Duszczyk, Gadamski, Danysz, & Lazarewicz, 2006). These data further support the feasibility of targeting elevations of intracellular calcium for neuroprotection.

Aequorin (AQ) is a CaBP isolated from the coelenterate *Aequorea Victoria*. AQ has been used for years as an indicator for monitoring calcium levels and has been shown to be safe and well tolerated by cells (Cobbold & Lee, 1991). AQ belongs to the EF-hand family of CaBPs, with EF-hand loops that are closely related to CaBPs in mammals (Toma et al., 2005). However, to date, no studies have investigated the therapeutic potential of AQ. Since HPC neurons exhibit profound aging-related decreases in CaBPs, we tested the hypothesis that replenishing CaBPs in the HPC will be neuroprotective when administered prior to an ischemic insult. The present studies were designed to test this hypothesis by injecting AQ directly into the HPC, inducing ischemia, and comparing the number of dead cells between AQ- and control-injected animals.

METHODS

Animals. Sixteen male F344 rats were used. Rats were housed individually on a 14/10 hour day/night light cycle with free access to food and water.

Surgery. Rats were anesthetized and mounted on a stereotaxic apparatus. Under aseptic conditions, the scalp was incised and retracted to the side, and the head was leveled between bregma and lambda. Each rat was prepared with bilateral stainless steel guide cannulae aimed at the dorsal hippocampus using sterotaxic coordinates (3.5 mm posterior, ± 2.6 mm lateral, 3.0 mm ventral) relative to bregma. Cannulae were secured to the skull with stainless steel screws and epoxy. A stainless steel cap remained in place when the rats were not being injected to prevent the guide cannulae from becoming occluded.

Drugs and Infusions. Rats were given an injection of either 4% AQ or aCSF 18-24 hours prior to decapitation. To facilitate neuronal uptake of AQ, 6% DMSO was added. All rats received bilateral infusions (0.5 µl/side) over 60 seconds and the injection cannulae remained in place for an additional 2 min to ensure diffusion. One hemisphere received AQ injections and the other aCSF injections. The injection cannulae were cut to extend 0.5 mm beyond the guide cannulae.

Slice Preparation. 400 µm thick slices were prepared using standard procedures (Moyer & Brown, 1998). Following slice recovery, in vitro ischemia was induced by transferring slices to fructose CSF (glucose replaced with fructose and bubbled with 95% N_2. The slices were in the ischemia solution for 5 minutes, and were then returned to oxygenated aCSF that contained 0.2% trypan blue for 30 minutes. Trypan blue readily penetrates dead cells and stains them blue while leaving living and healthy cells unstained (DeRenzis & Schechtman, 1973). The slices were rinsed in oxygenated room temperature aCSF twice then fixed in 10% neutral buffered formalin overnight in the refrigerator. The following day, the slices were cryoprotected, cut on a cryostat (40 µm) and mounted onto subbed slides.

Cell Counts. The slices were examined under an Olympus microscope (equipped with a digital camera) at 2X and 10X and pictures were taken. Trypan blue stained neurons within CA1 (about an 800 µm section at 10X) were counted. Statistical analyses were performed using Statview (v 5.0; SAS Institute, Inc., Cary, NC). An ANOVA was used to evaluate a drug and age effect.

Aequorin/Calbindin Labeling. Rats were anaesthetized and perfused with PBS followed by 10% neutral buffered formalin. Brains were post-fixed for 24-48 hours, cryoprotected with sucrose, and 50 µm sections were cut on a cryostat. Alternate sections were saved in PBS in 24-well culture plates. Slices were incubated in 1% $NaBH_4$ for 15 minutes, then washed with PBS for 10 minutes (two times). Slices were incubated in 10% NGS for 30 minutes, then in primary antibody solution (1:1000 of either mouse anti-calbindin [Sigma] or mouse anti-aequorin [Chemicon]) overnight at room temperature. The following day, the slices were washed in PBS for 10 minutes (two times) and incubated for 2 hours in secondary antibody solution in the dark (10 µg/mL of either anti-mouse Ig-Alexa 488 or anti-mouse Ig-Alexa 594[Molecular Probes]). The slices were then washed with PBS for 5 minutes (two times). They were mounted onto slides, coverslipped with Prolong Antifade, and sealed. The slices were examined and pictures were taken using an Olympus microscope equipped with a digital camera. Images were taken with the following objectives: 2X (0.06 NA), 10X (0.25 NA), and 20X (0.40NA).

RESULTS- EFFECTS OF AQ INJECTIONS ON CELL DEATH

1. Aequorin protects hippocampal neurons against ischemic cell death

A. Control

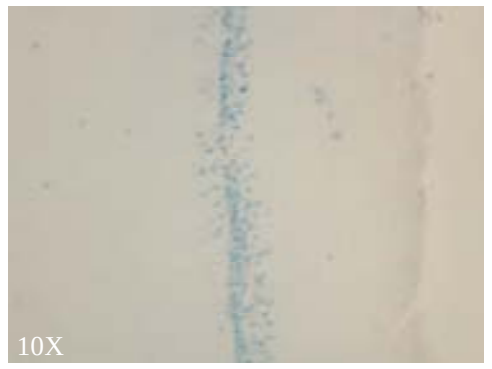
10X

B. Aequorin

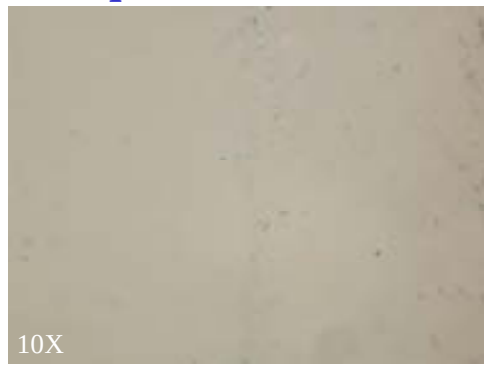
10X

C.

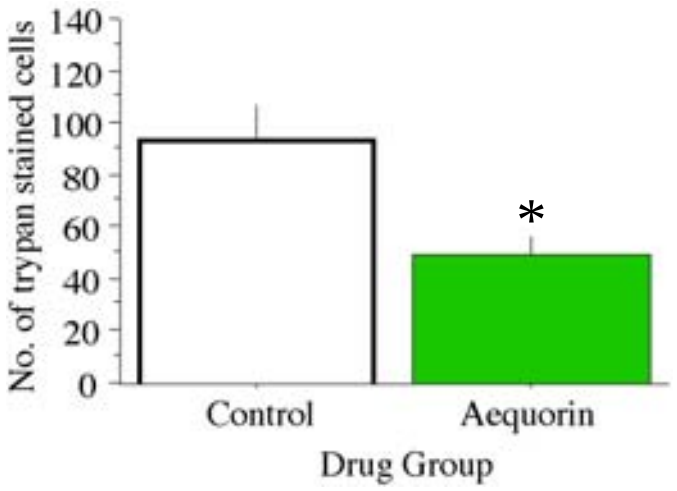
No. of trypan stained cells
140
120
100
80
60
40
20
0
*
Control
Aequorin
Drug Group

RESULTS- EFFECTS OF AGE AND AQ ON CELL DEATH

2. Aequorin protects neurons regardless of age

A. All slices treated individually

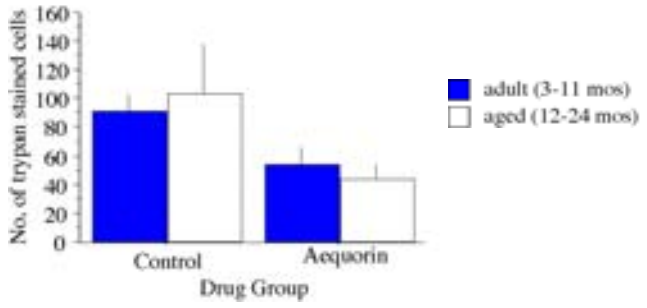
No. of trypan stained cells
160
140
120
100
80
60
40
20
0
Control
Aequorin
Drug Group
adult (3-11 mos)
aged (12-24 mos)

B. Slices averaged for each animal

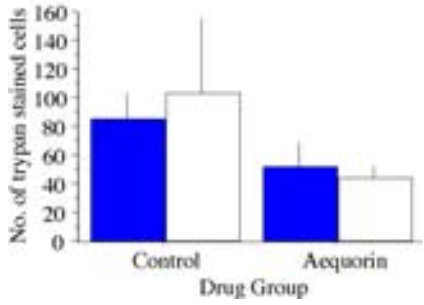
No. of trypan stained cells
160
140
120
100
80
60
40
20
0
Control
Aequorin
Drug Group

RESULTS- AQ LABELING

3. Aequorin injections in CA1 of hippocampus

A. AQ-injected

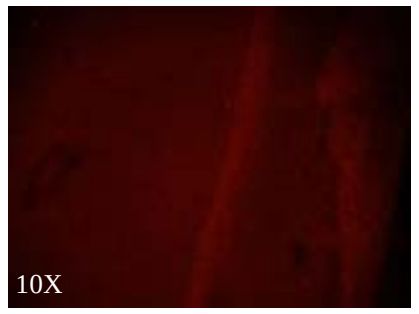
10X

B. control-injected

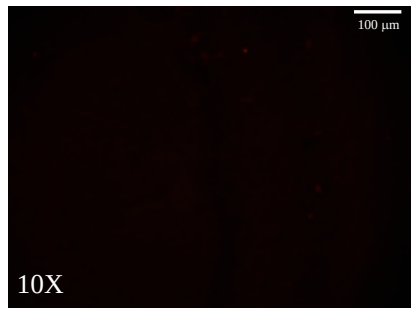
100 µm
10X

RESULTS- CANNULAE

4. Cannulae placement within CA1 of hippocampus

AP -3.5 mm

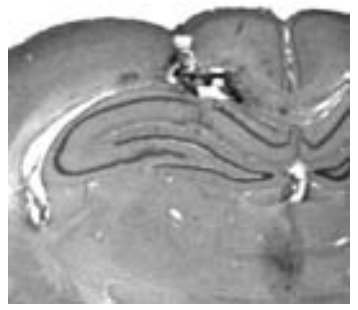

RESULTS- CALBINDIN LABELING

5. Decreased MTL calbindin staining with aging

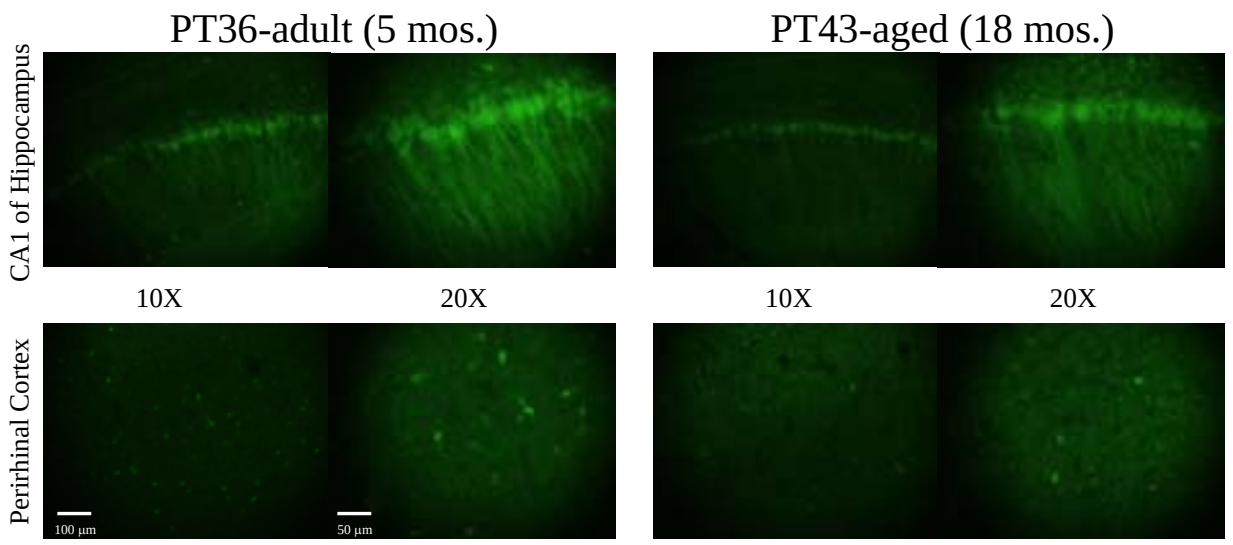
PT36-adult (5 mos.)
PT43-aged (18 mos.)
CA1 of Hippocampus
Perirhinal Cortex
10X
20X
10X
20X
100 µm
50 µm

SUMMARY

1. Injection of aequorin prior to ischemia is neuroprotective
 - *There were fewer dead cells in area CA1 of hippocampus in the aequorin-injected rats compared to control-injected animals*
2. The neuroprotective effect of aequorin occurs in both adult and aged rats
 - *Like the adult rats, aged rats had fewer dead cells in area CA1 of hippocampus after aequorin injections compared to control injections*
3. Aged rats exhibited less staining for the calcium binding protein calbindin
 - *This effect was observed in hippocampus and perirhinal cortex*
 - *A decrease in calbindin during aging may leave those neurons more vulnerable to aging-related neurodegenerative disorders*

REFERENCES

Baimbridge, K. G., Celio, M. R., & Rogers, J. H. (1992). Calcium-binding proteins in the nervous system. *Trends in Neuroscience, 15*(8), 303-308
Bano, D., Young, K. W., Guerin, C. J., Lefeuvre, R., Rothwell, N. J., Naldini, L., et al. (2005). Cleavage of the plasma membrane Na+/Ca2+ exchanger in excitotoxicity. *Cell, 120*(2), 275-285.
Chard, P. S., Bleakman, D., Christakos, S., Fullmer, C. S., & Miller, R. J. (1993). Calcium buffering properties of calbindin D28k and parvalbumin in rat sensory neurones. *Journal of Physiology, 472*, 341-357.
Choi, D. W. (1992). Excitotoxic cell death. *Journal of Neurobiology, 23*(9), 1261-1276.
Choi, D. W. (2005). Neurodegeneration: cellular defences destroyed. *Nature, 433*(7027), 696-698.
Cobbold, P. H., & Lee, J. A. C. (1991). Aequorin measurements of cytoplasmic free calcium. In P. H. Cobbold (Ed.), *Cellular calcium: a practical approach* (pp. 55-81). New York: Oxford University Press.
DeRenzis, F. A., & Schechtman, A. (1973). Staining by neutral red and trypan blue in sequence for assaying vital and nonvital cultured cells. *Stain Technology, 48*(3), 135-136.
De Jong, G. I., Naber, P. A., Van der Zee, E. A., Thompson, L. T., Disterhoft, J. F., & Luiten, P. G. M. (1996). Age-related loss of calcium binding proteins in rabbit hippocampus. *Neurobiology of Aging, 17*(3), 459-465.
Gant, J. C., Sama, M. M., Landfield, P. W., & Thibault, O. (2006). Early and simultaneous emergence of multiple hippocampal biomarkers of aging is mediated by Ca2+-induced Ca2+ release. *J Neurosci, 26*(13), 3482-3490.
Gary, D. S., Sooy, K., Chan, S. L., Christakos, S., & Mattson, M. P. (2000). Concentration- and cell type-specific effects of calbindin D28k on vulnerability of hippocampal neurons to seizure-induced injury. *Brain Research. Molecular Brain Research, 75*(1), 89-95.
Grotta, J. C. (1987). Current medical and surgical therapy for cerebrovascular disease. *N Engl J Med, 317*(24), 1505-1516.
Hof, P. R., & Morrison, J. H. (1991). Neocortical neuronal subpopulations labeled by a monoclonal antibody to calbindin exhibit differential vulnerability in Alzheimer's disease. *Experimental Neurology, 111*, 293-301.
Iacopino, A. M., & Christakos, S. (1990). Specific reduction of calcium-binding protein (28-kilodalton calbindin-D) gene expression in aging and neurodegenerative diseases. *Proceedings of the National Academy of Sciences (USA), 87*, 4078-4082.
Kristian, T., & Siesjo, B. K. (1998). Calcium in ischemic cell death. *Stroke, 29*(3), 705-718.
Krzywkowski, P., Potier, B., Billard, J. M., Dutar, P., & Lamour, Y. (1996). Synaptic mechanisms and calcium binding proteins in the aged rat brain. *Life Sciences, 59*(5/6), 421-428.
Lee, J. M., Zipfel, G. J., & Choi, D. W. (1999). The changing landscape of ischaemic brain injury mechanisms. *Nature, 399*(6738 Suppl), A7-14.
Lin, R. C., & Scheller, R. H. (2000). Mechanisms of synaptic vesicle exocytosis. *Annual Review of Cell and Developmental Biology, 16*, 19-49.
Makarewicz, D., Duszczyk, M., Gadamski, R., Danysz, W., & Lazarewicz, J. W. (2006). Neuroprotective potential of group I metabotropic glutamate receptor antagonists in two ischemic models. *Neurochem Int, 48*(6-7), 485-490.
Mattson, M. P., & Magnus, T. (2006). Ageing and neuronal vulnerability. *Nature Reviews. Neuroscience, 7*(4), 278-294.
Mikkonen, M., Alafuzoff, I., Tapiola, T., Soininen, H., & Niettinen, R. (1999). Subfield- and layer-specific changes in parvalbumin, calretinin and calbindin-D28K immunoreactivity in the entorhinal cortex in Alzheimer's disease. *Neuroscience, 92*(2), 515-532.
Moyer, J. R., Jr., & Brown, T. H. (1998). Methods for whole-cell recording from visually preselected neurons of perirhinal cortex in brain slices from young and aging rats. *Journal of Neuroscience Methods, 86*(1), 35-54.
Moyer, J. R., Jr., & Brown, T. H. (2006). Impaired trace and contextual fear conditioning in aged rats. *Behavioral Neuroscience, 120*(3), 612-624.
Sutherland, M. K., Wong, L., Somerville, M. J., Yoong, L. K. K., Bergeron, C., Parmentier, M., et al. (1993). Reduction of calbindin-28k mRNA levels in Alzheimer as compared to Huntington hippocampus. *Brain Research. Molecular Brain Research, 18*(1-2), 32-42.
Toma, S., Chong, K. T., Nakagawa, A., Teranishi, K., Inouye, S., & Shimomura, O. (2005). The crystal structures of semi-synthetic aequorins. *Protein Science, 14*(2), 409-416.
Villa, A., Podini, P., Panzeri, M. C., Racchetti, G., & Meldolesi, J. (1994). Cytosolic Ca^{2+} binding proteins during rat brain ageing: loss of calbindin and calretinin in the hippocampus, with no change in the cerebellum. *European Journal of Neuroscience, 6*, 1491-1499.
West, A. E., Chen, W. G., Dalva, M. B., Dolmetsch, R. E., Kornhauser, J. M., Shaywitz, A. J., et al. (2001). Calcium regulation of neuronal gene expression. *Proceedings of the National Academy of Sciences (USA), 98*(20), 11024-11031.
Yenari, M. A., Minami, M., Sun, G. H., Meier, T. J., Kunis, D. M., McLaughlin, J. R., et al. (2001). Calbindin d28k overexpression protects striatal neurons from transient focal cerebral ischemia. *Stroke, 32*(4), 1028-1035.

Supported by Quincy Biosciences, LLC